Poster # 569

In Vitro Resistance Selection Study and Favorable Human Pharmacokinetic Properties of RDEA427, a New HIV NNRTI

V Ong[1], B Groschel[1], Z Shen[1], R Straney[1], W Xu[1], Anneke Raney*[1], S Dueker[2], JL Girardet[1], K Manhard[1], and B Quart[1]
[1]Ardea Biosciences, Inc, San Diego, CA, US; [2]Vitalea Science, Davis, CA, US

Ardea Biosciences, Inc
4939 Directors Place
San Diego, CA 92121
www.ardeabio.com
araney@ardeabio.com
T: 858.652.6500 F: 858.625.0760

Abstract

Background: The emergence of resistance to non-nucleoside reverse transcriptase inhibitors (NNRTIs) limits the efficacy of these drugs. New NNRTIs with a higher genetic barrier to resistance and activity against these drug-resistant viruses are needed. RDEA427 exhibits excellent *in vitro* potency against wild-type (wt) and resistant viruses. The results of an *in vitro* resistance selection study and the favorable human pharmacokinetic (PK) properties of RDEA427 are presented.

Methods: Antiviral activities were determined using HIV-1 viruses containing wt or NNRTI-resistant sequences. HIV-1 virus resistant to RDEA427 was selected by serial passage in SupT1 cells. At each virus breakthrough, the drug concentration was doubled and the RT region was sequenced for genotypic analysis. Animal PK data were obtained following intravenous (IV) administration of RDEA427 and facilitated allometric scaling to predict its half-life in humans. Exposure and urinary excretion in humans were evaluated following IV administration of a microdose of [^{14}C]RDEA427. Quantitative analysis of [^{14}C]RDEA427 was performed using Accelerator Mass Spectrometry.

Results: RDEA427 had an EC_{50} value of ~1 nM and retained potent antiviral activity against NNRTI-resistant mutations such as K103N, Y188L, Y181C, K101E, K103N-Y181C, Y181C-G190A, and K103N-P225H. In an *in vitro* resistance-selection study, K103N virus was controlled by the initial RDEA427 concentration for at least 30 passages. Allometric scaling of clearance data from rat, dog, and monkey IV pharmacokinetic studies suggested RDEA427 possesses potential for once-a-day dosing. This was confirmed in humans, where RDEA427 displayed a mean half-life of ~41 hours and clearance of 0.24 L/hr/kg. The presence of an active metabolite, with antiviral activity equivalent to the parent and a half-life of ~50 hours, was also identified and quantified. Mean 24-hr urinary recovery of total radioactivity was 3.8% of the radioactive dose.

Conclusions: RDEA427 has shown potent *in vitro* activity against wild-type and NNRTI-resistant viruses. In a resistance-selection study, RDEA427 controlled K103N HIV-1 infection for more than 100 days, suggesting RDEA427 may provide a high barrier to resistance. Favorable PK of RDEA427 following IV microdosing in healthy volunteers supports the development of RDEA427, and the long half-life may mitigate the risk of a missed dose.

Introduction

The widespread use of NNRTIs in antiretroviral treatment regimens, while providing greater control of HIV infections, has caused the emergence of RT mutations conferring cross-resistance to most of the NNRTIs currently available. Some of these resistant mutations, especially K103N, have also been transmitted to antiretroviral treatment-naïve individuals. RDEA427, a new NNRTI in clinical development, has potent antiviral activity against wt HIV-1 and viruses with prevalent NNRTI-resistant mutations. It also retains activity against many of the recently identified etravirine resistance associated mutations (RAMs). RDEA427 shows a lower potential for CYP3A4 induction than etravirine and TMC278 and better metabolic stability and lower covalent binding than TMC278.[1] An *in vitro* selection study with RDEA427 in K103N and Y181C HIV-1-infected SupT1 cells suggests RDEA427 will have a genetic barrier to resistance similar to or better than etravirine and TMC278. In a human PK microdose study, RDEA427 had a mean half-life of 41 hours, compatible with once daily dosing.

Figure 1. RDEA427 activity against HIV-1 viruses containing prevalent NNRTI-resistant mutations

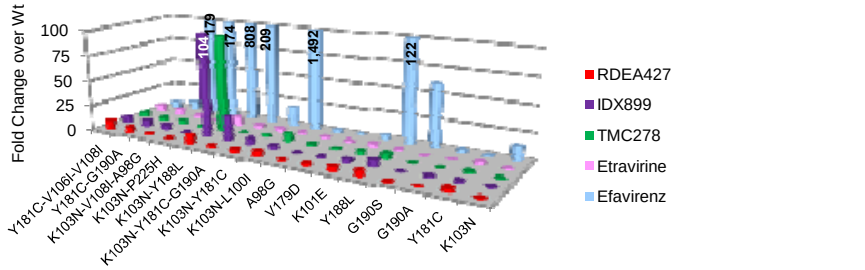
Fold Change over Wt
100
75
50
25
0
104
179
174
808
209
1,492
122
Y181C-V106I-V108I
Y181C-G190A
K103N-V108I-A98G
K103N-P225H
K103N-Y188L
K103N-Y181C-G190A
K103N-Y181C
K103N-L100I
A98G
V179D
K101E
Y188L
G190S
G190A
Y181C
K103N
RDEA427
IDX899
TMC278
Etravirine
Efavirenz

- **RDEA427 has excellent activity (FC<1) against the K103N mutant virus, the most commonly transmitted resistant virus in antiretroviral naïve patients[2]**
- **RDEA427 retains antiviral activity against viruses with other prevalent efavirenz and etravirine resistant mutations**

Methods and Results

Figure 2. *In vitro* selection time course in SupT1 cells infected with K103N and Y181C NL4-3 HIV-1

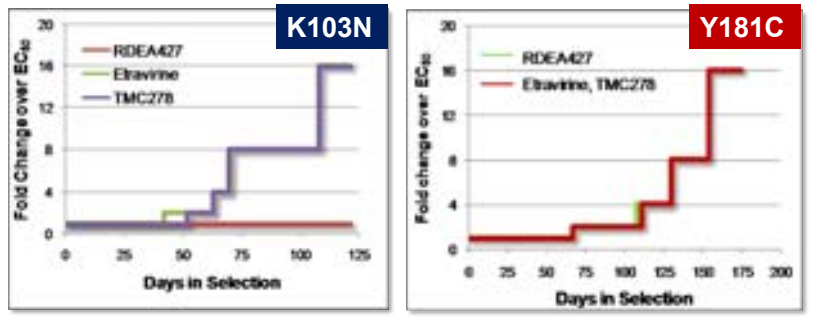
K103N
RDEA427
Etravirine
TMC278
Fold Change over EC_{50}
Days in Selection
Y181C
RDEA427
Etravirine, TMC278
Fold change over EC_{50}
Days in Selection

- **RDEA427 maintained control over K103N HIV-1 *in vitro* longer than etravirine and TMC278**
- **Y181C HIV-1 broke through RDEA427, etravirine and TMC278 suppression at the same rate**

Table 1. Mutations selected *in vitro* by RDEA427, etravirine and TMC278 in K103N and Y181C HIV-1-infected SupT1 cells

	K103N HIV-1		Y181C HIV-1		
Compound	Etravirine	TMC278	RDEA427	Etravirine	TMC278
Passage #	P30	P30	P49	P49	P49
Concentration	8 x EC_{50}	8 x EC_{50}	16 x EC_{50}	16 x EC_{50}	16 x EC_{50}
RT Mutations	(K103N), L100I	(K103N), L100I	(Y181C), V179T, G333R	(Y181C), P14A, V106I, V179I, Q222Q/H, F227F/C	(Y181C), V106I, H221H/Y, A272A/E, V314V/I, E399E/G

- **In K103N virus-infected cells, L100I emerged by passage 30 in the presence of both etravirine and TMC278; the virus had not broken through RDEA427 by P30.**
- **RDEA427-treated Y181C virus cultures contained V179T, which shows reduced susceptibility to etravirine. In Y181C virus-infected cells, mutations V179I and P14A were selected by etravirine, and V106I was selected by etravirine and TMC278 by passage 31.**

RDEA427 Human PK Prediction:
Using an allometric scaling approach, total clearance (CL) and volume of distribution (Vd) parameters were estimated from animal plasma concentration-time data from corresponding intravenous administrations. From these parameters, the half-life ($t_{1/2}$) in human was projected. The allometric equation is as follows:

$$Y = \alpha\cdot(BW)^{\beta} \quad \text{or} \quad \log Y = \alpha + \beta\cdot\log BW$$

where **Y** is CL or Vd, **α** is the allometric coefficient, **β** is the allometric exponent, and **BW** is the body weight.

Figure 3. Clearance (CL) and volume (Vd) correlations from rat, dog, and monkey IV dosing

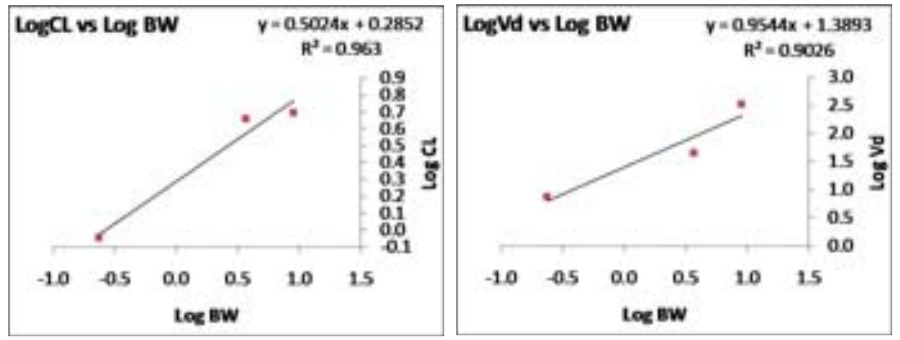
LogCL vs Log BW
y = 0.5024x + 0.2852
R^2 = 0.963
Log CL
Log BW
LogVd vs Log BW
y = 0.9544x + 1.3893
R^2 = 0.9026
Log Vd
Log BW

Table 2. PK parameters for animals (actual) and human (projected)

Species	CL (L/hr)	CL (L/hr/kg)	Vd (L)	Vd (L/kg)	$t_{1/2}$ (hr)
Rat	0.895	3.58	7.38	29.5	4.40
Monkey	4.56	1.24	44.1	12.0	7.89
Dog	4.98	0.553	327	36.3	5.06
Human (projected)	*16.3*	*0.233*	*1413*	*20.2*	*60.1*

Table 3. Observed human PK data from RDEA427 IV microdose study

	C_0 (pg/mL)	AUC_{last} (hr·pg/mL)	AUC_{INF} (hr·pg/mL)	CL (L/hr)	Vd (L)	$t_{1/2}$ (hr)
Mean	**1166**	**4321**	**5593**	**19.1**	**1036**	**41.1**
SD	663	845	1441	6.46	267	18.3
CV%	56.9	19.6	25.8	33.8	25.8	44.6

- **Four subjects were administered 100 μg [^{14}C]RDEA427 by intravenous injection in 4 mL solution as a slow bolus. Plasma exposure and urinary excretion of [^{14}C]RDEA427 and metabolite were evaluated by LC/AMS.**

Figure 4. Plasma concentration-time profiles from human IV microdose

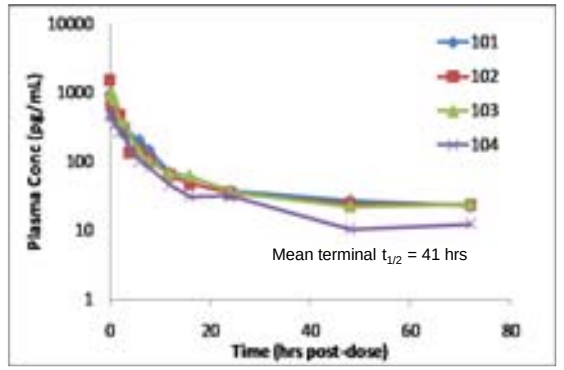
Plasma Conc (pg/mL)
Time (hrs post-dose)
101
102
103
104
Mean terminal $t_{1/2}$ = 41 hrs

- **Similar plasma concentration-time profiles were observed for the four subjects**
- **The mean terminal half-life of RDEA427 was 41 hours**

Figure 5. LC-AMS metabolite profiling: Plasma concentration-time profiles of RDEA427 and its main metabolite from human microdose

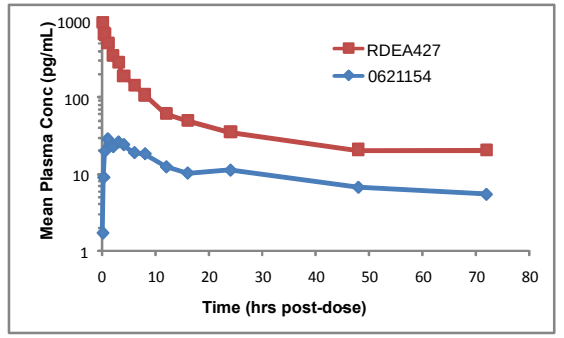
Mean Plasma Conc (pg/mL)
Time (hrs post-dose)
RDEA427
0621154

- **The metabolite, 0621154, showed a $T_{1/2}$ of ~50 hours and represented ~17% of parent AUC in the RDEA427 microdose study**
- **No adverse events and no clinically relevant abnormalities in laboratory tests were observed during the microdose study**

Figure 6. Fold Change in activity of RDEA427 metabolite 0621154 against prevalent NNRTI-resistant viruses

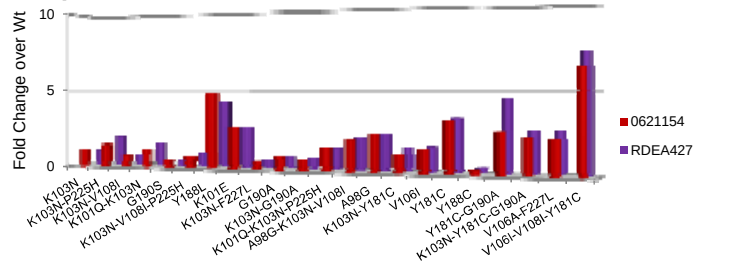
Fold Change over Wt
K103N
K103N-P225H
K103N-V108I
K101Q-K103N
G190S
K103N-V108I-P225H
Y188L
K101E
K103N-F227L
G190A
K103N-G190A
K101Q-K103N-P225H
A98G-K103N-V108I
A98G
K103N-Y181C
V106I
Y181C
Y188C
Y181C-G190A
K103N-Y181C-G190A
V106A-F227L
V106I-V108I-Y181C
0621154
RDEA427

- **The main RDEA427 metabolite observed, 0621154, has an EC_{50} of 1.1 nM against wt HIV-1 and activity equivalent to RDEA427 in NNRTI-resistant viruses**

Conclusions

- RDEA427 is active against NNRTI-resistant mutant viruses, including prevalent transmitted viruses and etravirine RAMs.
- In vitro selection showed longer suppression of K103N virus by RDEA427 than etravirine and TMC278, suggesting a higher resistance threshold against this virus.
- Allometric scaling from rat, dog and monkey projected human clearance and half-life similar to actual values in IV human microdose experiment.
- RDEA427 has a half-life of 41 hours after the IV microdose, which would support once daily dosing.
- An active metabolite of RDEA427, 0621154, with equivalent activity to RDEA427 in wild-type and NNRTI-resistant viruses, displays a ~ 50-hour half-life.
- With its excellent activity against resistant virus, long plasma half-life and lower potential for drug interactions, RDEA427 appears to be a good candidate for further development.

Acknowledgment: We thank Joanne Graham for her expert technical assistance.

References

1. Raney A, Hamatake R, Xu W, *et al.* (2008) 15th Conference on Retroviruses and Opportunistic Infections (CROI), Boston, MA.
2. Smith D, Moini N, Pesano R, *et al. (2007)* Clinical utility of HIV standard genotyping among antiretroviral-naïve individuals with unknown duration of infection. Clin Infect Dis ;44:456-8.

The 16th Conference on Retroviruses and Opportunistic Infections (CROI 2009), February 8-11, 2009, Montréal, Canada