Resistance to RDEA806 Requires Multiple Mutations Which Have Limited Cross-Resistance to Other NNRTIs

Poster # H-1222

W. Xu, B. Groschel, R. Straney, Z. Zhang, D. Bellows, R. Hamatake, J-L. Girardet, B. Quart, A. Raney
Ardea Biosciences, Inc.

Ardea Biosciences, Inc.
4939 Directors Place
San Diego, CA 92121
www.ardeabio.com
Tel: 858-622-6500
Fax: 858-625-0760

Abstract

BACKGROUND: RDEA806 is a novel non-nucleoside reverse transcriptase inhibitor (NNRTI) with potent *in vitro* activity against wild-type and NNRTI-resistant HIV-1. Phase 2a clinical trial data demonstrated robust antiviral activity in naïve patients. RDEA806 suppressed viral breakthrough in an *in vitro* resistance selection study for 9 months, indicating a high genetic barrier to resistance. The genotypic and phenotypic analyses of the mutant viruses selected by RDEA806 in this resistance selection study will be presented.

METHODS: Selection of HIV-1 virus resistant to RDEA806 was performed in infected SupT1 cells. The mutations identified in the RT region at the time of virus breakthrough for each drug concentration were engineered into a wild-type vector. Activity was measured using the VSV-G protein pseudotyped NL4-3.Luc.R-E- virus containing the mutation(s).

RESULTS: RDEA806 concentration was increased to 1500 nM in the virus culture over the course of 13 months. The first consensus mutation selected, K104E, was identified after > 300 days in culture and showed essentially no loss of susceptibility to RDEA806. Other NNRTIs tested also retained maximal activity against this mutant virus. RDEA806 showed a minor loss of activity to the next virus selected, K104E-E138K-T240I, while efavirenz retained full activity. Virus with the ultimate combination contained 5 RT mutations, K104E-E138K-T240I-V179D-F227L, and had very low replication capacity. While RDEA806 lost potency against this virus, other NNRTIs showed only minor to moderate cross resistance.

CONCLUSIONS: Prolonged suppression of viral breakthrough in this resistance selection study suggests that RDEA806 has a high genetic barrier to resistance. Phenotypic analysis of selected mutations indicates that multiple mutations are necessary for loss of susceptibility to RDEA806. Minor cross-resistance to some of the mutation patterns is observed by some of the NNRTIs tested.

Introduction

The development of resistance to the most widely used non-nucleoside reverse transcriptase inhibitors (NNRTIs) limits the efficacy of these drugs. New NNRTIs with higher genetic barriers to resistance and activity against viruses with common drug-resistant mutations are needed for the effective treatment of patients carrying these mutations. RDEA806 is a novel NNRTI with *in vitro* activity across a broad range of NNRTI-resistant viruses and robust anti-HIV activity in infected patients. The study presented here suggests that RDEA806 may also provide a higher threshold to resistance, with the potential for more durable suppression of the virus in patients.

Methods

Selection and determination of RDEA806-resistant (RDEA806^R) mutations. SupT1 cells were infected with wild-type (wt) or K103N HIV-1 in the presence of the EC_{50} concentration of RDEA806 or efavirenz (EFV). At each virus breakthrough (massive syncytia formation), the concentration of inhibitor was doubled. Genotypic analysis of the emerging viruses was performed by amplification of the pro-viral DNA and subsequent sequencing of the RT region in ≥ 10 individual PCR clones recovered at each breakthrough point.

Construction and testing of NNRTI-resistant viruses. Clinically prevalent NNRTI-resistant mutations and the RDEA806^R mutations were constructed into the HIV-1 molecular clone pNL4-3.Luc.R-E-. A VSV-G protein-pseudotyped luciferase-expressing HIV-1 was used to determine the antiviral activities. The EC_{50} values were determined by non-linear regression using XLfit4.

Results

Figure 1. RDEA806 Activity Against Prevalent NNTRI-resistant Viruses

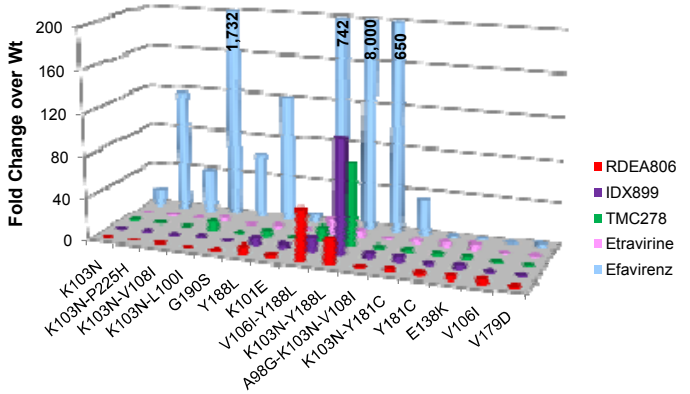
Fold Change over Wt
200
160
120
80
40
0
1,732
742
8,000
650
K103N
K103N-P225H
K103N-V108I
K103N-L100I
G190S
Y188L
K101E
V106I-Y188L
K103N-Y188L
A98G-K103N-V108I
K103N-Y181C
Y181C
E138K
V106I
V179D
RDEA806
IDX899
TMC278
Etravirine
Efavirenz

- RDEA806 has broad spectrum activity against NNRTI-resistant viruses
- RDEA806 has excellent activity (FC <1) against the K103N mutation, the most commonly transmitted resistant virus in antiretroviral naïve patients (up to 11.7%)[1]
- The resistance profile of RDEA806 is superior to that of EFV and similar to those of other NNRTIs

Figure 2. Time Course of Viral Breakthrough

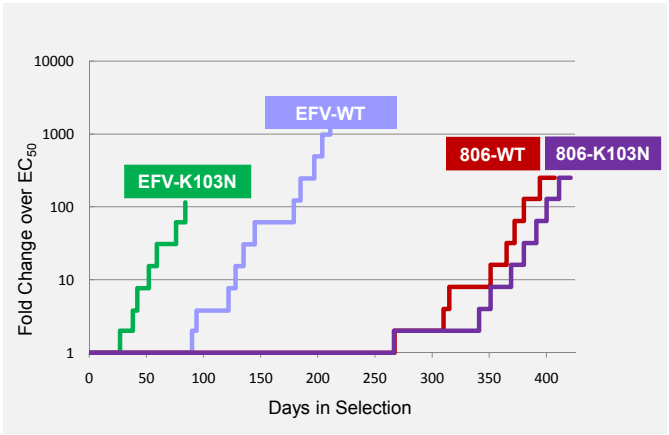
Fold Change over EC_{50}
10000
1000
100
10
1
0
50
100
150
200
250
300
350
400
Days in Selection
EFV-WT
EFV-K103N
806-WT
806-K103N

- RDEA806 provides more durable suppression of WT and K103N virus than EFV
- WT HIV-1 required 267 days of culturing in RDEA806 at the EC_{50} level to break through, compared with 90 days in EFV
- RDEA806 required multiple nucleotide changes for high level resistance

Figure 3. Mutants Selected by RDEA806 in Wild-type Virus

Consensus Genotype			K104E		K104E E138K T240I	K104E E138K	K104E E138K T240I	K104E E138K T240I V179D	K104E E138K T240I V179D F227L	
Total Days			310	315	351	365	372	380	394	407
Fold EC_{50}			2	4	8	16	32	64	128	250
RT aa #	Wt	Mut								
75	V	L								
104	K	E								
138	E	K								
179	V	D								
227	F	L								
240	T	I								
# of clones			11	10	10	10	10	10	10	10

- K104E was the first mutation to emerge, followed gradually by E138K, T240I, V179D and F227L

Figure 4. NNRTI Activity Against RDEA806-selected Mutant Viruses: WT HIV-1

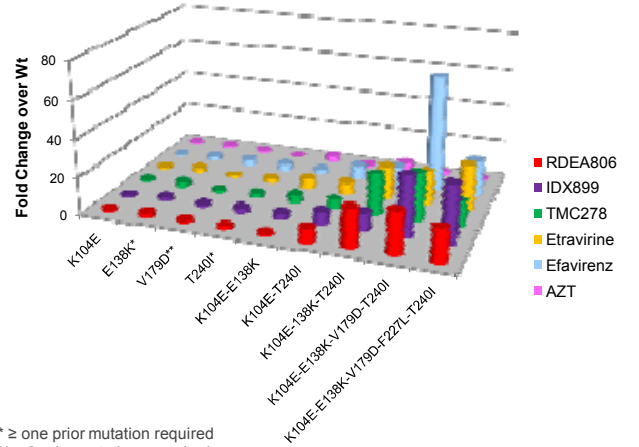
Fold Change over Wt
80
60
40
20
0
K104E
E138K*
V179D**
T240I*
K104E-E138K
K104E-T240I
K104E-138K-T240I
K104E-E138K-V179D-T240I
K104E-E138K-V179D-F227L-T240I
RDEA806
IDX899
TMC278
Etravirine
Efavirenz
AZT

* ≥ one prior mutation required
** ≥ 3 prior mutations required

- RDEA806 requires at least 3 mutations for > 10x loss in activity
- Other NNRTIs are active against most of the RDEA806-selected mutant viruses
- Viruses with ≥ 3 mutations have very low replication capacity

Figure 5. Mutants Selected by RDEA806 in K103N Virus

Consensus Genotype			K103N	K103N K512E	K103N V106A			K103N V106A K219E			K103N V106A K219E K512E
Total Days			267	341	351	369	380	391	400	411	421
Fold EC_{50}			1	2	4	8	16	32	64	128	250
RT aa #	Wt	Mut									
103	K	N									
106	V	A									
219	K	E									
227	F	L									
512	K	E									
# of Clones			21	10	10	10	10	10	12	10	10

- Starting with K103N, selection of predominant mutations was slow; V106A was followed by K219E, K512E and F227L

Figure 6. NNRTI Activity Against RDEA806-selected Mutant Viruses: K103N HIV-1

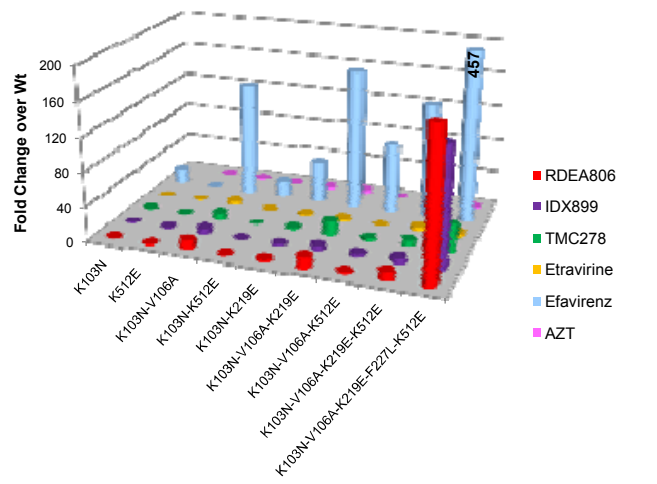
Fold Change over Wt
200
160
120
80
40
0
457
K103N
K512E
K103N-V106A
K103N-K512E
K103N-K219E
K103N-V106A-K219E
K103N-V106A-K512E
K103N-V106A-K219E-K512E
K103N-V106A-K219E-F227L-K512E
RDEA806
IDX899
TMC278
Etravirine
Efavirenz
AZT

- RDEA806 requires at least 4 mutations for significant loss in activity
- Other NNRTIs remain active against the RDEA806-selected mutant viruses, with TMC278 and etravirine effective against the most resistant virus
- Viruses with ≥ 3 mutations have very low replication capacity

Figure 7. RDEA806 Demonstrated Excellent Viral Load Suppression with No Genotypic Changes in a Phase 2a Monotherapy Trial

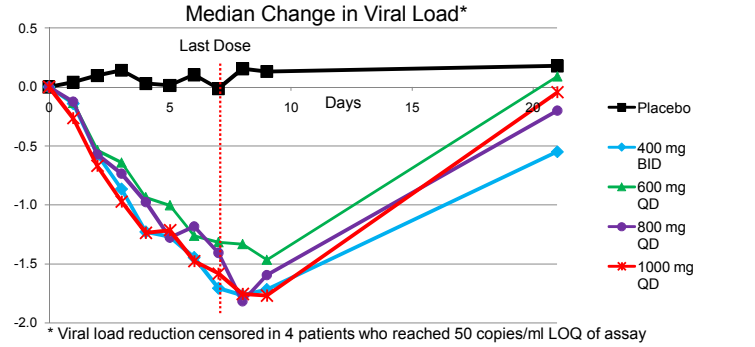
Median Change in Viral Load*
0.5
0.0
-0.5
-1.0
-1.5
-2.0
Last Dose
0
5
10
15
20
Days
Placebo
400 mg BID
600 mg QD
800 mg QD
1000 mg QD

* Viral load reduction censored in 4 patients who reached 50 copies/ml LOQ of assay and several patients started on triple therapy after Day 10 and prior to follow-up visit

- In 36 patients treated with RDEA806, 144 samples were analyzed for RT genotype in the Virco®TYPE HIV-1 assay.
- No genotypic changes characteristic of known NNRTI resistance or of the mutations identified in the *in vitro* resistance study were observed in 36 patients at the end of treatment or two weeks after the last dose.

Conclusions

- In this long-term selection study, RDEA806 provided more durable suppression against Wt and K103N viruses than efavirenz.
- The pathways to RDEA806 resistance were different for Wt and K103N viruses.
- Other than efavirenz activity on the K103N-containing viruses, most of the viruses selected remained susceptible to other NNRTIs.
- The viruses engineered with the mutation patterns that showed high levels of resistance replicated very poorly.
- The prolonged suppression of viral breakthrough and the requirement for multiple amino acid changes for high level resistance suggest that RDEA806 has a high genetic barrier to resistance.
- Consistent with this *in vitro* study, no genotypic or phenotypic changes occurred during RDEA806-201, a Phase 2a monotherapy study in treatment-naïve HIV-infected individuals (ICAAC Oral Presentation #4707)

Reference
1. Smith D, Moini N, Pesano R, Little S, et al. Clinical utility of HIV standard genotyping among antiretroviral-naïve individuals with unknown duration of infection. Clin Infect Dis 2007;44:456-8.

Disclosures
W. Xu, Z. Zhang, D. Bellows, R. Hamatake are former employees of Ardea Biosciences. B. Groschel, R. Straney, J.-L. Girardet, B. Quart and A. Raney are current employees of Ardea Biosciences.

48th Annual ICAAC / IDSA 46th Annual Meeting, 25-28 October, 2008, Washington, D.C., USA