The Selective MEK Inhibitor RDEA119: Synergy with Multiple Classes of Anti-Cancer Agents

Poster # 3700

M. Chapman, C. Iverson, M. Miampamba, *C. Yu, *A. Adjei, J-L. Girardet, B. Quart, J. Miner

[1]Ardea Biosciences, Inc and *Roswell Park Cancer Institute

Ardea Biosciences, Inc
4939 Directors Place
San Diego, CA 92121
www.ardeabio.com

Abstract

The RAS-RAF-MEK-ERK pathway is an attractive target for cancer therapeutics because of its role in both proliferation as well as cell death. Indeed, members of this pathway are among the most frequently mutated genes in cancer, and include clear examples of 'oncogene addiction', such as constitutively active B-Raf mutations commonly seen in malignancies including melanoma, colorectal cancer, NSCLC, and thyroid cancer. RDEA119 is a highly specific, allosteric MEK1/2 inhibitor currently in early clinical trials in advanced cancer patients as both a monotherapy and in combination with the multiple kinase inhibitor sorafenib. RDEA119 shows broad promise in oncology due to its efficacy in cell proliferation and apoptosis assays, multiple human xenograft models in mice, and its favorable pharmacokinetics. We present data demonstrating that RDEA119 synergizes strongly with sorafenib in cell based systems, as demonstrated by isobolographic analysis, as well as in xenograft models. The mechanism of synergy between RDEA119 and sorafenib is examined using selective compounds which inhibit only a subset of the kinases targeted by sorafenib. We suggest that inhibition of B-Raf by sorafenib plays a role in the synergy. Additionally, we will present combination studies with RDEA119 and several classes of emerging anti-cancer agents, including standard chemotherapeutic agents such as gemcitabine, as well as drugs targeting the PI3K pathway. These synergistic interactions provide supporting evidence for the use of combinations of these agents and RDEA119 in clinical settings.

Introduction

Simple inhibition of the RAF-MEK-ERK pathway will likely prove to be an effective treatment in some cancer patients, given the role of activation of this pathway in many tumors. In such cases, success will depend on potent and specific MEK1/MEK2 inhibition in the tumor. In other situations, the combination of a MEK inhibitor with other anti-cancer agents (either cytotoxic chemotherapeutics or more targeted agents) may have added benefit. We have tested RDEA119 in combination with several existing and emerging cancer drugs in an effort to evaluate the potential for additive and synergistic effects. The combination of RDEA119 and sorafenib acts synergistically in many preclinical systems, and is currently being evaluated in a Ph1/2 trial in advanced cancer patients. Other combinations also show promise, for example RDEA119 combined with either mTOR inhibitors, EGFR inhibitors, or gemcitabine.

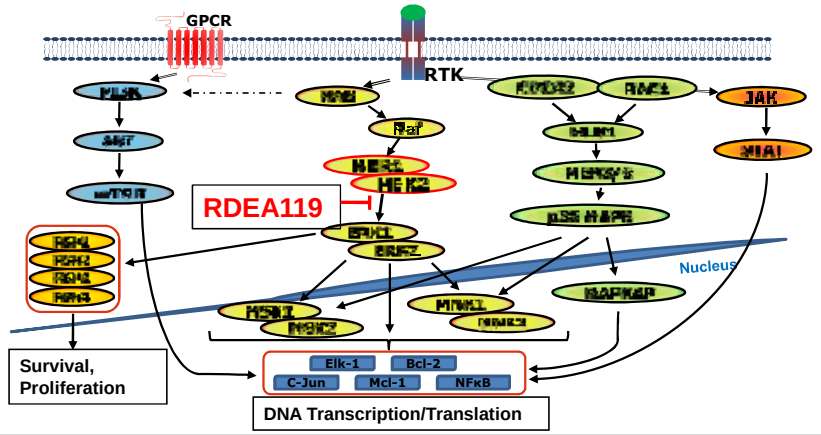

GPCR
RTK
JAK
Raf
RDEA119
Nucleus
Survival, Proliferation
Elk-1
Bcl-2
C-Jun
Mcl-1
NFκB
DNA Transcription/Translation

Results

RDEA119 and Sorafenib Synergistically Inhibit Proliferation

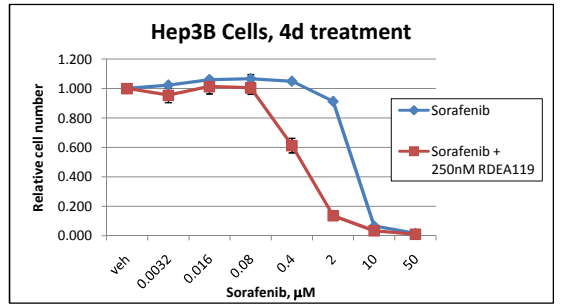

Hep3B Cells, 4d treatment
Relative cell number
Sorafenib, µM
Sorafenib
Sorafenib + 250nM RDEA119

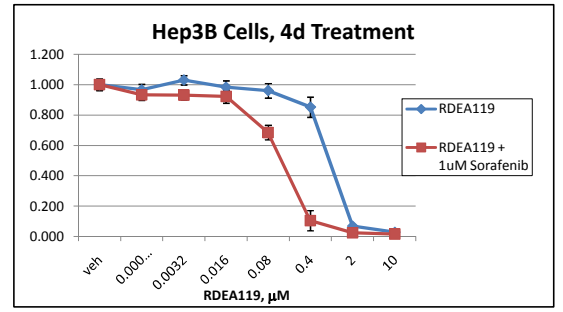

Hep3B Cells, 4d Treatment
RDEA119, µM
RDEA119
RDEA119 + 1uM Sorafenib

Hep3B cells were treated as indicated for 4 days and then cell number was assayed using Cell Titer Glo. Each point represents the mean of triplicate wells -/+ SEM. Results are representative of multiple experiments

Synergy in Multiple Cell Types

Cell line	Cancer type	Known Mutational Status	RDEA119 Growth IC50, uM	Synergy?
MiaPaCa2	Pancreatic	K-Ras	0.15	+
A549	NSCLC	K-Ras	0.59	
HCT-116	CRC	K-Ras, PI3KCA	0.20	
NCI-H460	NSCLC	K-Ras, PI3KCA	3.63	
A375	Melanoma	B-Raf	0.08	
Colo205	CRC	B-Raf	0.06	
MCF-7	Breast	PI3KCA	0.85	
Hop-92	NSCLC	-	0.05	+
Huh7	Hepatoma	-	1.50	+
MV522	NSCLC	-	0.02	+
U2-OS	Osteosarcoma	-	0.30	+
Hep3B	Hepatoma	-	0.10	+
D37	Glioma	-	0.50	+

Cell lines were assessed as above (or by isobolographic analysis).

Xenograft Results

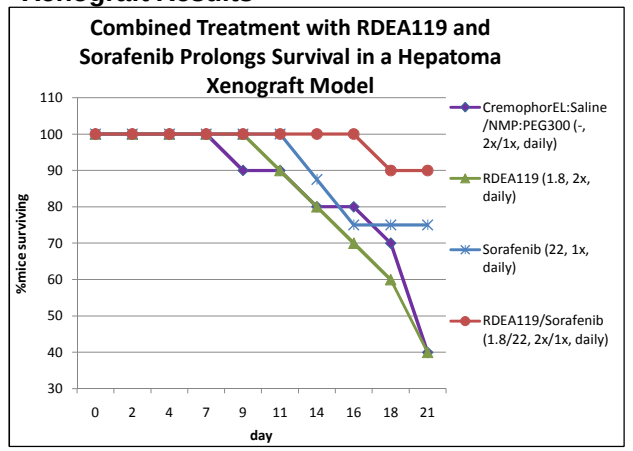

Combined Treatment with RDEA119 and Sorafenib Prolongs Survival in a Hepatoma Xenograft Model
%mice surviving
day
CremophorEL:Saline /NMP:PEG300 (-, 2x/1x, daily)
RDEA119 (1.8, 2x, daily)
Sorafenib (22, 1x, daily)
RDEA119/Sorafenib (1.8/22, 2x/1x, daily)

Female Balb/c nu/nu mice were implanted s.c. with 2.5 million Huh7 cells. Treatment began 21 days later when the average tumor size reached $300mm^3$. Animals were culled during the study if tumors reached $2000mm^3$. Treatments were generally well tolerated.

Inhibition of Raf by Sorafenib may play a Role in RDEA119 + Sorafenib Synergy

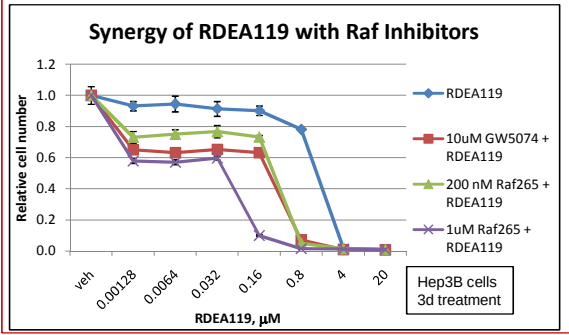

Synergy of RDEA119 with Raf Inhibitors
Relative cell number
RDEA119, µM
RDEA119
10uM GW5074 + RDEA119
200 nM Raf265 + RDEA119
1uM Raf265 + RDEA119
Hep3B cells 3d treatment

- Sorafenib inhibits VEGFR, EGFR, cKit, and Raf kinase activity.
- Sunitinib, which inhibits VEGFR, EGFR, cKit, and Flt3, but not Raf, does not synergize with RDEA119 in cell lines where sorafenib does (data not shown).
- Raf inhibitors GW5074 and Raf265 both synergistically inhibit Hep3B cell proliferation in combination with RDEA119.
- This suggests that Raf inhibition in combination with MEK1/2 inhibition may be beneficial as an anti-cancer therapy in certain tumors.

Synergy Screen for RDEA119 over Multiple Drug Categories

			Sunitinib	Erlotinib	Gefitinib	Lapanitib	Imatinib	Everolimus	Temsirolimus	Vorinostat	Gemcitabine	Temozolomide
Cell line	Tumor origin	Known mutational status	VEGFR, EGFR, cKit, Flt-3	EGFR tk	EGFR tk	EGFR, HER2	BCR-Abl, cKit	mTOR	mTOR	HDAC	nucleoside analog	alkylator
Colo205	Colorectal cancer	B-Raf										
G361	Melanoma	B-Raf										
MDA-MB231	Breast cancer	B-Raf K-Ras										
MiaPaCa-2	Pancreatic Cancer	K-Ras										
A549	NSCLC	K-Ras										
Panc1	Pancreatic cancer	K-Ras										
SW403	Colorectal Cancer	K-Ras										
AGS	Gastric cancer	K-Ras PIK3CA										
HCT-116	Colorectal Cancer	K-Ras PIK3CA										
MCF-7	Breast cancer	PIK3CA										
SW579	Thyroid cancer	-										
Hep3B	Hepatoma	-										
786-O	Renal carcinoma	-										

Synergy seen in at least 4 different dose combinations | Synergy seen in at least 1 dose combination | No evidence of synergy | Not tested

For most combinations shown, RDEA119 and the indicated drug were tested for inhibition of proliferation in a matrix format for synergy analysis. Isobolographic analysis was performed, and the results categorized as shown. Significant antagonism was rarely seen. Drug combination matrices were tested at least twice, with consistent results.

- Synergy with EGFR inhibitors was observed in several K-Ras mutant cell lines.
- mTOR inhibitors appear to synergize with RDEA119 in PIK3CA mutant lines by blocking both pathways downstream of Ras.
- Synergy with gemcitabine occurs in several cell lines and is under further investigation.

RDEA119 Synergizes with Gemcitabine in Several Cancer Cell Lines

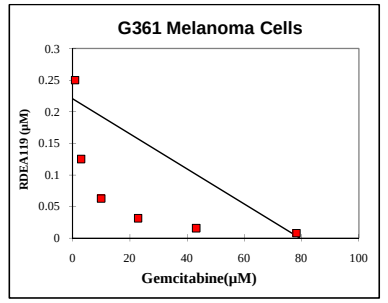

G361 Melanoma Cells
RDEA119 (µM)
Gemcitabine(µM)

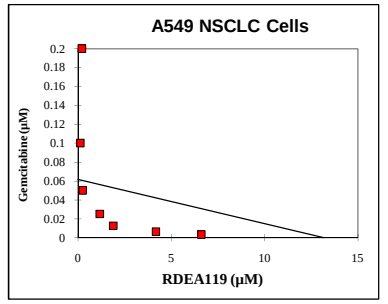

A549 NSCLC Cells
Gemcitabine (µM)
RDEA119 (µM)

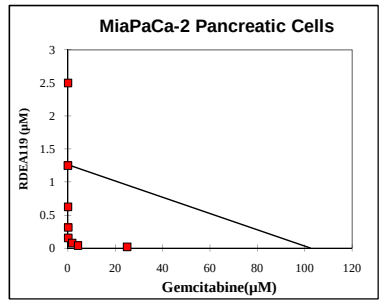

MiaPaCa-2 Pancreatic Cells
RDEA119 (µM)
Gemcitabine(µM)

Isobolograms showing synergy of RDEA119 and gemcitabine in inhibiting proliferation of three different cell lines. Points below the isobole indicate a synergistic interaction.

RDEA119 Synergizes with the mTOR Inhibitor Everolimus in a Colon Cancer Line Carrying K-Ras and PIK3CA Mutations

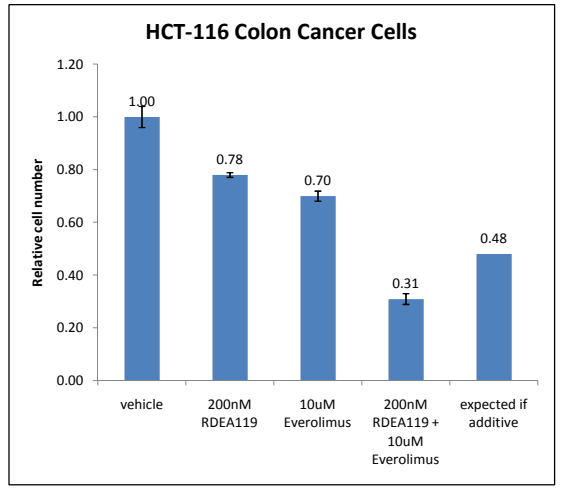

HCT-116 Colon Cancer Cells
Relative cell number
1.00
0.78
0.70
0.31
0.48
vehicle
200nM RDEA119
10uM Everolimus
200nM RDEA119 + 10uM Everolimus
expected if additive

HCT-116 cells were treated as indicated for 5 days and then cell number was assayed using Cell Titer Glo. Each point represents the mean of triplicate wells -/+ SEM. Results are representative of multiple experiments

Conclusions

- RDEA119, a potent and selective inhibitor of MEK1/2, is progressing through a Phase 1 trial in advanced cancer patients. We demonstrated the potential for synergistic combinations of RDEA119 with other anti-cancer agents.
- RDEA119 shows strong synergy with sorafenib across many cell lines, as well as in preclinical models (see also poster #3692, C. Yu et al). This combination is currently being evaluated in a clinical trial.
- Inhibition of Raf kinase by sorafenib may play a role in the synergy seen with RDEA119.
- Several other drugs, when combined with RDEA119, behave synergistically in blocking cell proliferation. Further evaluation of these interactions is in progress.

AACR 100th Annual Meeting . April 18-22, 2009, Denver , Colorado